4907

Siponimod First-Dose Effects in Patients With SPMS Receiving Concomitant Selective Serotonin Reuptake Inhibitor Therapy

Amos Katz,[1] Bruce A C Cree,[2] Le H Hua,[3] Derrick Robertson,[4] Brandon Brown,[5] Desiree Dunlop,[5] Xiangyi Meng,[5] Wendy Su,[5] Amit Bar-Or[6]

[1]Linda Cardinale MS Center, Freehold, NJ; [2]Department of Neurology, UCSF Weill Institute for Neurosciences, University of California San Francisco, San Francisco, CA; [3]Lou Ruvo Center for Brain Health, Las Vegas, NV; [4]University of South Florida, Multiple Sclerosis Center, Tampa, FL; [5]Novartis Pharmaceuticals Corporation, East Hanover, NJ; [6]Center for Neuroinflammation and Experimental Therapeutics and Department of Neurology, Perelman School of Medicine, University of Pennsylvania, Philadelphia, PA

Introduction

- Siponimod (Mayzent®) is a selective sphingosine 1-phosphate receptor ($S1P_1$ and $S1P_5$) modulator, approved for the treatment of adults with relapsing forms of multiple sclerosis (MS), including clinically isolated syndrome, relapsing-remitting MS and active secondary progressive MS (SPMS)[1]
- In EXPAND, a phase 3 trial examining the efficacy and safety of siponimod in an SPMS population, siponimod significantly reduced risk of 3 month and 6 month confirmed disability progression versus placebo[2]
- Some selective serotonin reuptake inhibitors (SSRIs), such as citalopram and escitalopram, may cause corrected QT (QTc) interval prolongation and torsades de pointes[3]
- Given that transient heart rate decrease at treatment initiation is an expected pharmacodynamic effect of S1P receptor modulators, it is important to understand the cardiac effects in patients receiving concomitant SSRIs
- First-dose observation with siponimod is only required in certain cardiac conditions, but it is important to understand the cardiac effects in patients receiving concomitant SSRIs

Objective

- Evaluate first-dose effects of siponimod in patients receiving concomitant SSRIs during the phase 3 EXPAND trial

Methods

- Analysis included all participants who had been randomized to siponimod in EXPAND
- EXPAND was a phase 3, 36 month, randomized, placebo-controlled trial of siponimod 2 mg/day in adults (18-60 years old) with SPMS, Expanded Disability Status Scale (EDSS) score of 3.0-6.5, and EDSS progression in the 2 years before study[2]
- The following patient subgroups randomized to siponimod in EXPAND were assessed:
 - any concomitant SSRI at first dose (Day 1)
 - either concomitant citalopram or escitalopram at first dose (Day 1)
- Assessments were made for frequency and severity of treatment-emergent adverse events (TEAEs) and serious TEAEs
- Vital signs (sitting mean pulse rate and blood pressure) were recorded hourly after first dose
- Additional cardiac assessments include:
 - dose initiation sitting mean pulse (summary of change from pre-dose by hour on Day 1)
 - bradyarrhythmias on Day 1
 - proportion of patients requiring extended monitoring
- Individuals were discharged after a 6 hour observation period unless they had a heart rate lower than 45 beats per minute (bpm), QTc interval of at least 500 ms, new-onset second-degree or higher atrioventricular (AV) block, or the investigator determined that the patient had symptoms associated with reduced heart rate. Patients meeting these criteria were required to undergo extended observation

Results

Demographics

- Of 1105 patients randomized to siponimod in EXPAND, on Day 1:[2]
 - 1099 were included in analyses[2]
 - 5 did not receive study drug
 - 1 failed to supply a signed consent form
 - 167 received any concomitant SSRI
 - 85 received concomitant citalopram or escitalopram
- Baseline demographics in the subgroups of patients receiving siponimod with any SSRI or with citalopram/escitalopram on Day 1 were similar to those in all patients receiving siponimod (**Table 1**)

Table 1. Baseline demographics

	Siponimod (all) N=1105	Siponimod with any SSRI[a] N=167	Siponimod with citalopram/escitalopram[a] N=85
Sex, n (%)			
Female	669 (60.5)	119 (71.3)	64 (75.3)
Male	436 (39.5)	48 (28.7)	21 (24.7)
Age, years, mean (SD)	48.0 (7.84)	49.2 (7.02)	48.3 (6.86)
Age group, years, n (%)			
18-30	26 (2.4)	2 (1.2)	1 (1.2)
31-40	162 (14.7)	17 (10.2)	9 (10.6)
41-55	716 (64.8)	117 (70.1)	65 (76.5)
Over 55	201 (18.2)	31 (18.6)	10 (11.8)
Weight, kg, mean (SD)	71.53 (15.685)	73.70 (17.274)	73.32 (17.988)
Body mass index, kg/m², mean (SD)	24.90 (4.842)	26.05 (5.521)	25.90 (5.499)
Race, n (%)			
Asian	31 (2.8)	0	0
Black or African American	7 (0.6)	3 (1.8)	1 (1.2)
Other	12 (1.1)	0	0
Unknown	5 (0.5)	0	0
White	1050 (95.0)	164 (98.2)	84 (98.8)
Duration of MS since first symptoms, mean (SD)	17.12 (8.385)	18.20 (8.220)	18.09 (8.205)
Number of relapses in past year, mean (SD)	0.2 (0.54)	0.2 (0.49)	0.2 (0.57)
Number of relapses in past 2 years, mean (SD)	0.7 (1.20)	0.5 (1.04)	0.6 (1.20)

[a]On Day 1
MS, multiple sclerosis; SD, standard deviation; SSRI, selective serotonin reuptake inhibitor

Table 2. Cardiac treatment-emergent adverse events on Day 1

	Siponimod (all) N=1099	Siponimod with any SSRI N=167	Siponimod with citalopram/escitalopram N=85
Any SAE, n (%)	4 (0.4)	0	0
Bradycardia	2 (0.2)	0	0
Atrioventricular block second degree	1 (0.1)	0	0
Heart rate decreased	1 (0.1)	0	0
Pyelonephritis chronic	1 (0.1)	0	0
Any AE, n (%)	199 (18.1)	36 (21.6)	21 (24.7)
Bradycardia	29 (2.6)	3 (1.6)	2 (2.4)
Sinus bradycardia	6 (0.5)	0	0
Atrioventricular block first degree	4 (0.4)	0	0
Heart rate decreased	4 (0.4)	1 (0.6)	1 (1.2)
Atrioventricular block second degree	3 (0.3)	0	0
Electrocardiogram QTc interval prolonged	3 (0.3)	3 (1.6)	1 (1.2)

AE, adverse event; SAE, serious AE; SSRI, selective serotonin reuptake inhibitor

Figure 1. Occurrence of bradyarrhythmias at Day 1 after first dose

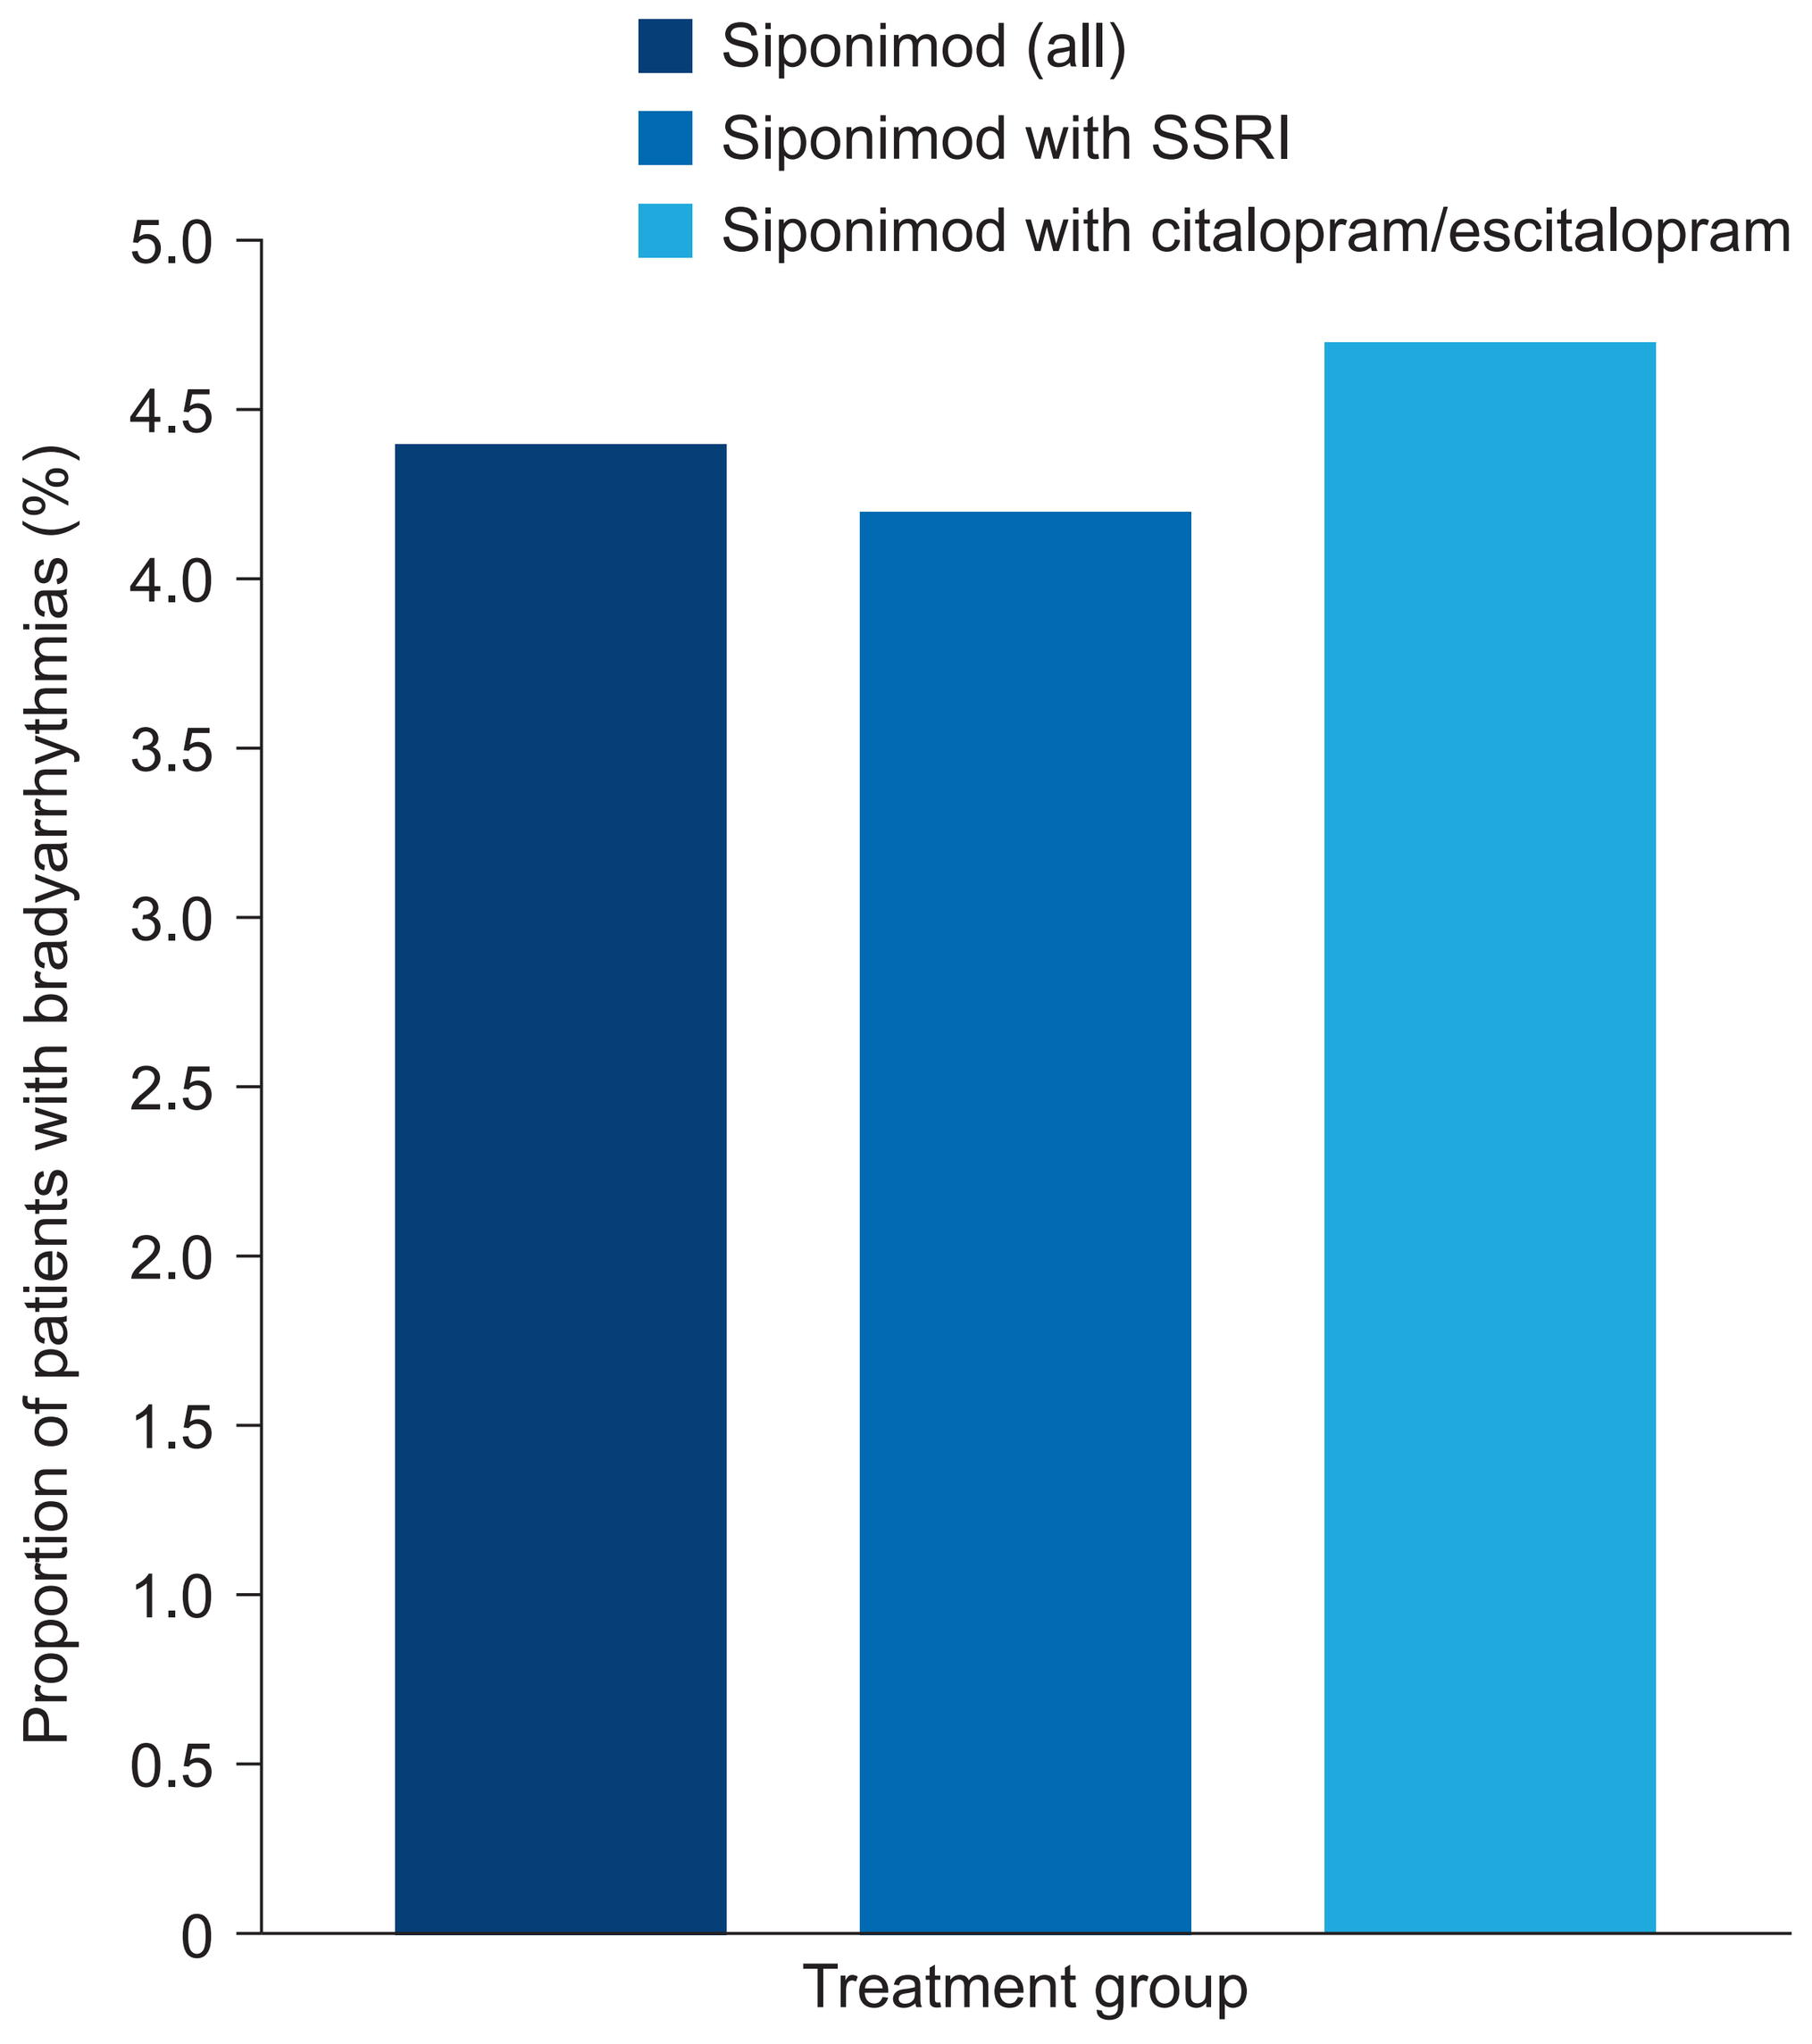

Siponimod (all)
Siponimod with SSRI
Siponimod with citalopram/escitalopram
Proportion of patients with bradyarrhythmias (%)
5.0
4.5
4.0
3.5
3.0
2.5
2.0
1.5
1.0
0.5
0
Treatment group

Siponimod, N=1099; SSRI, N=167; citalopram/escitalopram, N=85
SSRI, selective serotonin reuptake inhibitor

Figure 2. Change in mean sitting heart rate in 6 hours following first dose

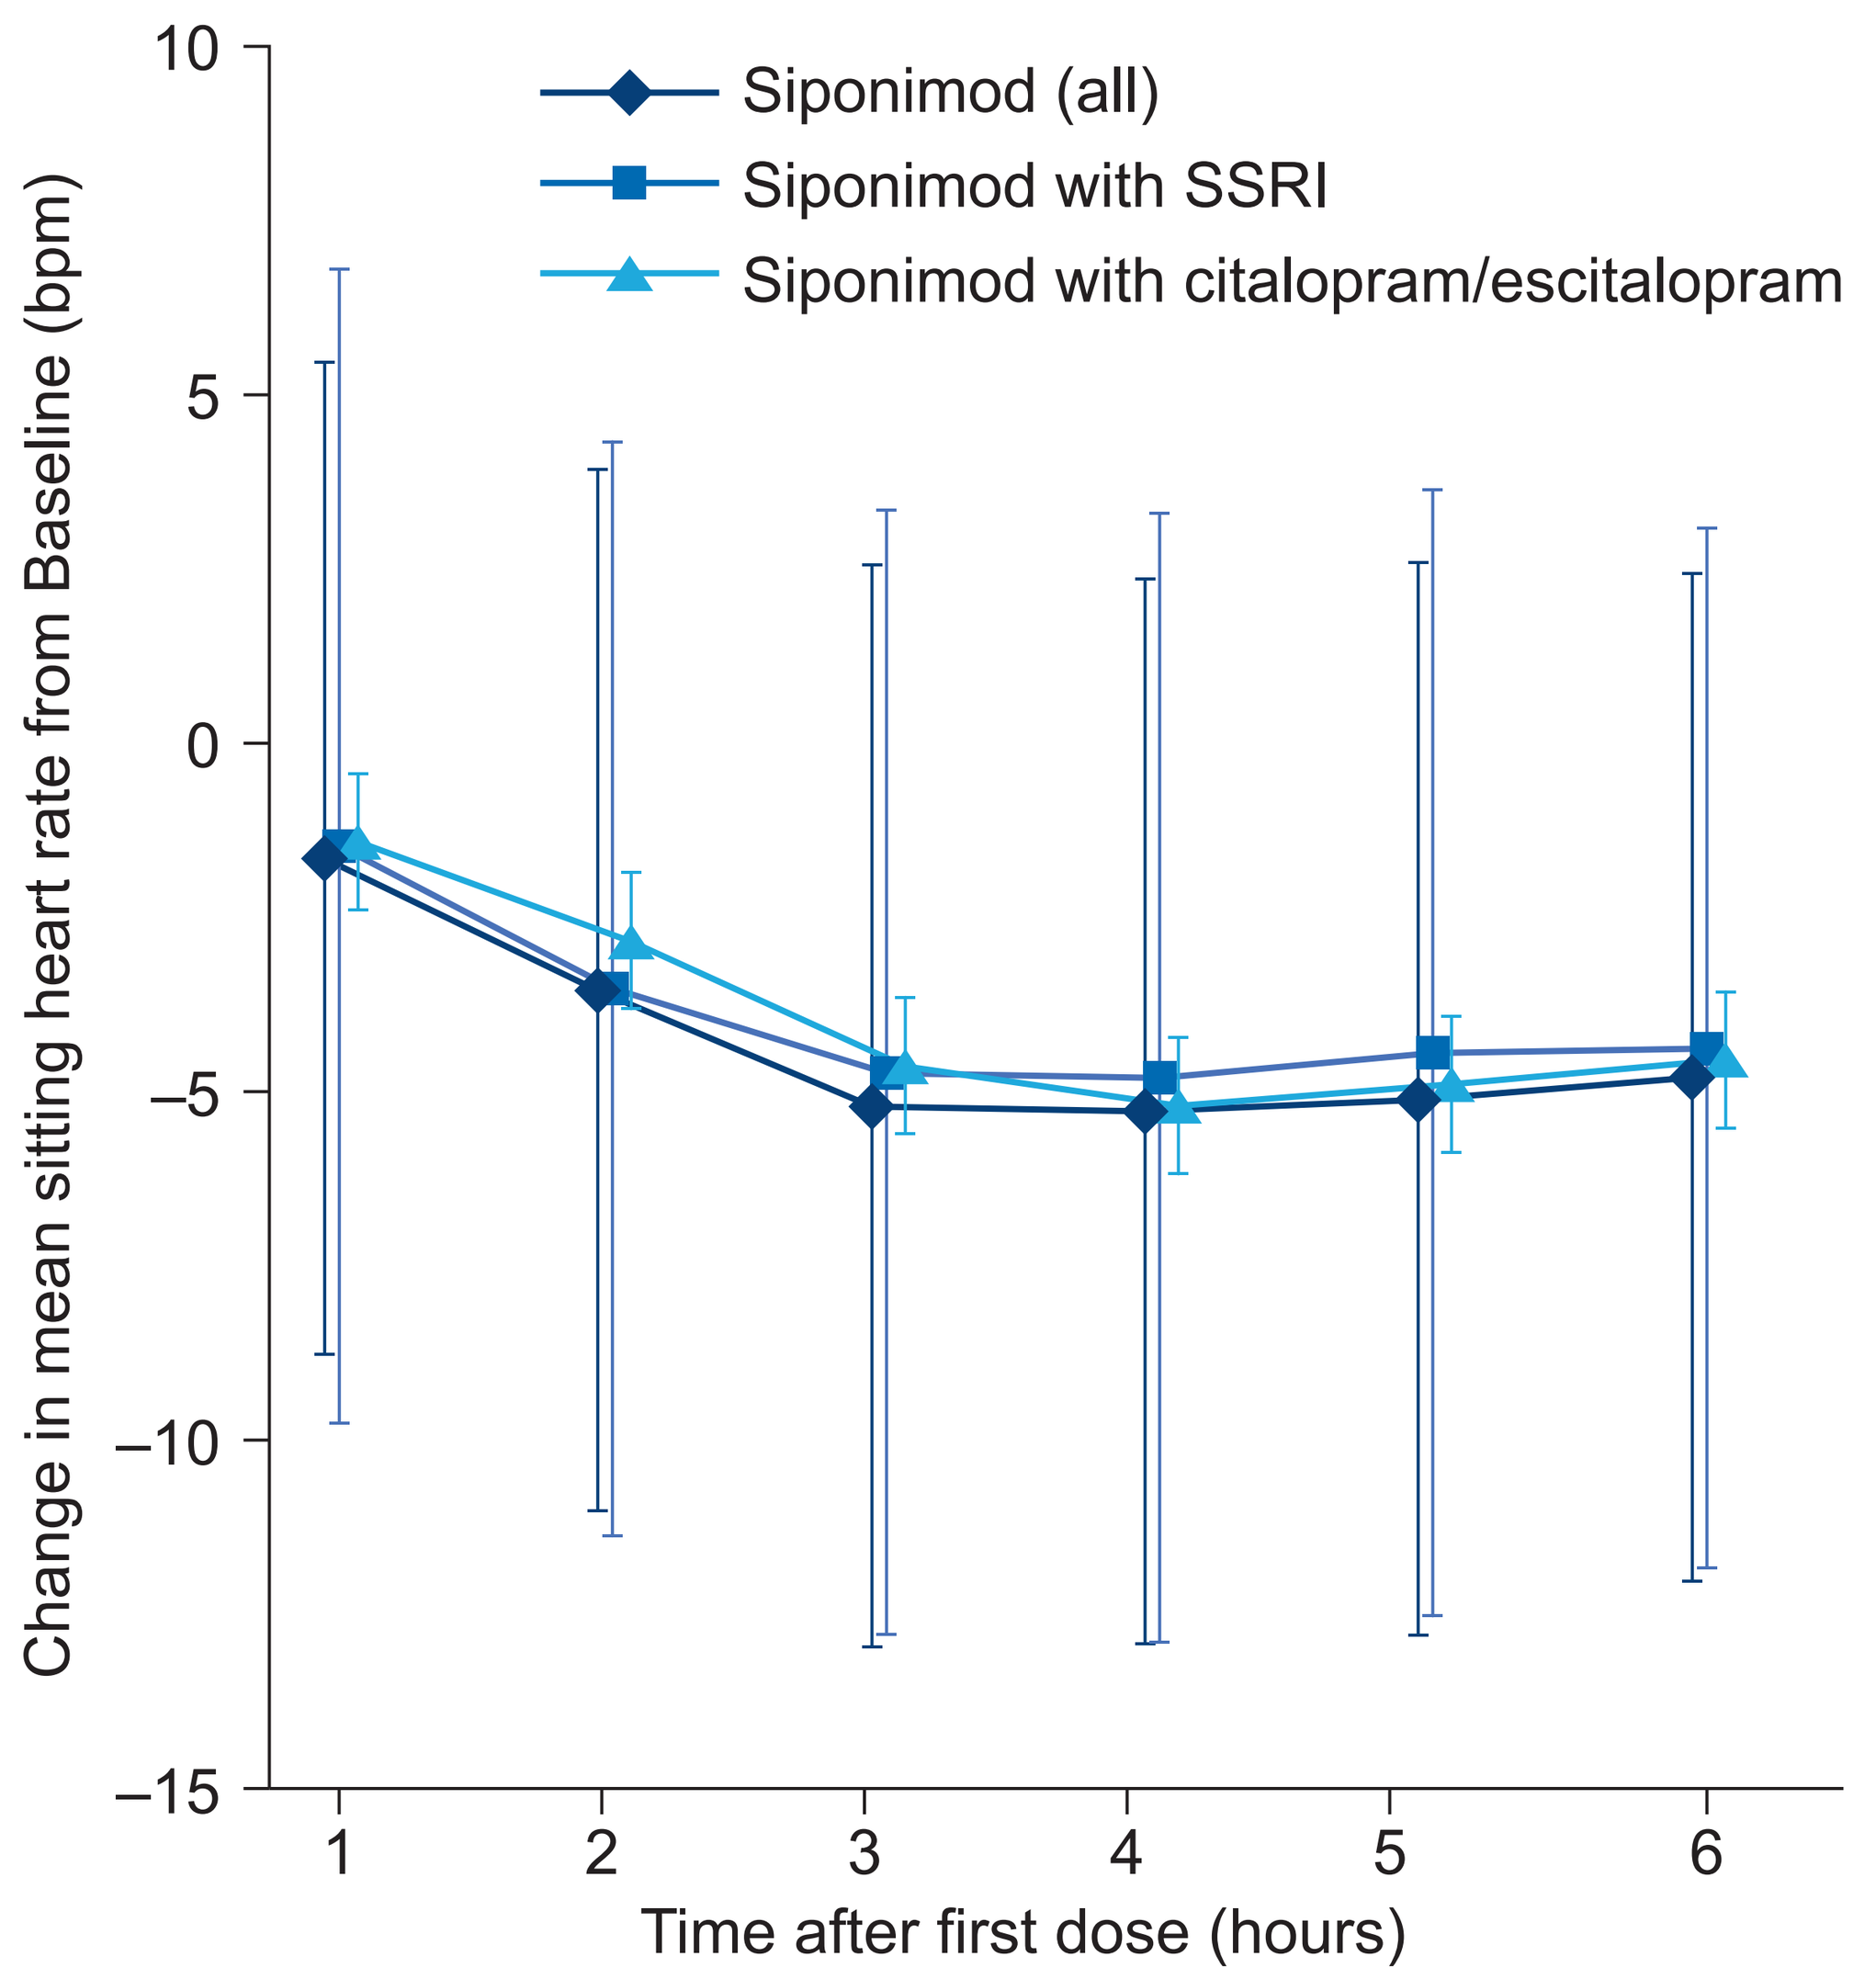

Siponimod (all)
Siponimod with SSRI
Siponimod with citalopram/escitalopram
Change in mean sitting heart rate from Baseline (bpm)
10
5
0
−5
−10
−15
1
2
3
4
5
6
Time after first dose (hours)

Siponimod, N=1099; SSRI, N=167; citalopram/escitalopram, N=85
Data points represent change from Baseline in sitting heart rate (bpm), mean and standard deviation
bpm, beats per minute; SSRI, selective serotonin reuptake inhibitor

Clinical experience

- Most patients did not require extended monitoring and were discharged at 6 hours post first dose
 - Proportions of patients requiring extended monitoring were similar in all patients receiving siponimod (8.9%; 86 of 962), patients receiving siponimod with any SSRI (8.6%; 13 of 152) and those receiving siponimod with citalopram/escitalopram (10.4%; 8 of 77)
- On Day 1 after first dose, four patients (0.4%) receiving siponimod had serious AEs (SAEs), two (0.2%) had bradycardia and one (0.1%) had second-degree AV block
 - None of these occurred in the subgroups receiving siponimod with SSRI or with citalopram/escitalopram (**Table 2**)
- Few patients receiving siponimod had cardiac TEAEs on Day 1 (**Table 2**)
 - Bradycardia: 2.6%
 - First-degree AV block: 0.4%
 - Second-degree AV block: 0.3%
 - Prolonged QTc interval: 0.3%
- Incidence of cardiac AEs was low in the subgroups receiving siponimod with SSRI or with citalopram/escitalopram (**Table 2**)
 - Bradycardia: 1.8% and 2.4%, respectively
 - Prolonged QTc interval: 1.8% and 1.2%, respectively
 - There were no reported incidents of first- or second-degree AV block
- Of all patients receiving siponimod (with or without SSRI), three patients (0.3%) discontinued drug owing to first- or second-degree AV block or bradycardia. No patients receiving concomitant SSRIs, including citalopram and escitalopram, had a cardiac AE causing treatment discontinuation

Cardiac effects post dose

- Bradyarrhythmias occurred in 48 of 1099 patients (4.4%) receiving siponimod (with or without SSRIs), 7 of 167 patients (4.2%) in the concomitant SSRI subgroup and 4 of 85 patients (4.7%) in the concomitant citalopram/escitalopram subgroup (**Figure 1**)
- Lowest mean sitting heart rate in the concomitant SSRI and concomitant citalopram/escitalopram subgroups was reached at 4 hours post dose
 - Mean change from Baseline: −4.82 and −5.22 bpm, respectively (**Figure 2**), and recovery had begun by 5 hours
 - This was consistent with that observed in all patients receiving siponimod (−5.30 bpm)

Conclusions

- Concomitant SSRI use on Day 1 did not appear to affect cardiac outcomes or heart rate associated with siponimod treatment initiation

References

1. Novartis Pharmaceuticals Corporation. Prescribing information. MAYZENT® 2019. Available from: https://www.novartis.us/sites/www.novartis.us/files/mayzent.pdf (Accessed June 26, 2020).
2. Kappos L, *et al. Lancet.* 2018;391:1263-1273.
3. Bermel RA, *et al. Mult Scler Rel Dis.* 2015;4:273-280.

Acknowledgments

The authors wish to thank all patients who participated in the EXPAND study. Editorial support was provided by Grace Jeong, PhD of Alphabet Health, New York, NY, USA, which was funded by Novartis Pharmaceuticals Corporation. This poster was previously presented at the Consortium of Multiple Sclerosis Centers (CMSC) Virtual Annual Meeting 2020. The final responsibility for the content lies with the authors.

Disclosures

Amos Katz has received consulting and speaker fees for Biogen, Genentech, Novartis and Sanofi. **Bruce AC Cree** has received personal compensation in the past 36 months for consulting from Akili, Alexion, Atara, Biogen, EMD Serono, Novartis, Sanofi and TG Therapeutics. **Le H Hua** has received speaker, advisory board and consulting fees from Biogen, Celgene, EMD Serono, Genentech, Genzyme and Novartis. **Derrick Robertson** is a consultant for Alexion, Biogen, Celgene, EMD Serono, Genentech, Novartis, Sanofi Genzyme and Teva Neuroscience; is on a speaker bureau for Acorda, Alexion, Biogen, Celgene, EMD Serono, Genentech, Mallinckrodt, Novartis, Sanofi Genzyme and Teva Neuroscience; and has received grant support from Actelion, Biogen, EMD Serono, Genentech, Mallinckrodt, MedDay, MedImmune, Novartis, PCORI, Sanofi Genzyme and TG Therapeutics. **Brandon Brown, Desiree Dunlop, Xiangyi Meng**, and **Wendy Su** are employees of Novartis Pharmaceuticals Corporation. **Amit Bar-Or** has received personal compensation for consulting, serving on scientific advisory boards or speaking activities, or a combination thereof, from Biogen Idec, F. Hoffmann-La Roche, Immunic, Genentech, GlaxoSmithKline, Merck Serono, Novartis and Sanofi Genzyme.

© 2021 Novartis Pharmaceuticals Corporation.

Poster Presentation at the American Academy of Neurology (AAN) Virtual Annual Meeting, 2021.

This study was sponsored by Novartis Pharmaceuticals Corporation, East Hanover, NJ, USA.

Text: Q3a9ea
To: 8NOVA (86682) US Only
+18324604729 North, Central and South Americas; Caribbean; China
+447860024038 UK, Europe and Russia
+46737494608 Sweden, Europe
Visit the web at:
http://novartis.medicalcongressposters.com/Default.aspx?doc=3a9ea

Scan this QR code to download a copy of this poster

Copies of this poster obtained through QR (Quick Response) code are for personal use only and may not be reproduced without written permission of the authors

Presenter email address: akatzmd@gmail.com